Increasing Representation and Diversity in Clinical Trials of Alzheimer's Disease: Recruitment of Ethnically Diverse Participants With Alzheimer's Disease in the Phase 1 ASCENT Clinical Trials of PRX012

P044

Chad Swanson, PhD[1,*]; Ryan E. Tooker, PhD[1]; Donna Masterman, MD, MS[1]; Courtney Fitzgerald, BS[1]; Ann Johnson, MS[1]; Lingnan Li, PhD[1]; Mary E. Quiceno, MD, FAAN[1]; Hideki Garren, MD, PhD[1]; Gene G. Kinney, PhD[1]

[1]Current and former employees of Prothena Biosciences Inc, Brisbane, CA, USA

BACKGROUND

- Alzheimer's disease (AD) is a chronic, progressive neurodegenerative disease defined by the presence of amyloid pathology.[1] It is characterized by impacts on multiple cognitive domains, development of behavioral and neuropsychiatric symptoms, and impaired day-to-day functional abilities.[1,2] Recent evidence demonstrates the slowing of clinical decline with plaque-clearing anti-amyloid beta (Aβ) monoclonal antibodies (mAbs), supporting a causative role for amyloid in AD.[3,4]
- Historically, populations participating in clinical trials of AD have been predominantly White older adults,[5-7] whereas Black and Hispanic/Latino older adults appear more likely to have AD or other dementias than White older adults.[8] Disparities in ethnic and racial representation can put into question whether clinical trial findings apply to all at-risk populations.[5-8]
- Historical comparators may help contextualize the issue of diversity of clinical trials in AD:
 - A systematic review of 49 global randomized controlled trials in AD reported that a "striking minority" of the overall population was Black or Hispanic/Latino (4.4% cumulatively).[6]
 - Another systematic review of 101 global AD trials suggests that the high proportion of White participants in AD trials (~95%) has not meaningfully changed over time.[7]
 - Of 2021 US enrollees in Medicare Part A and/or B, ~10% were Hispanic/Latino and ~11% were Black.[9]
- The low proportion of ethnic and racial diversity in clinical trials of AD was seen in the US participants of the TRAILBLAZER-ALZ 2 donanemab Phase 3 trial, in which 6% of participants were Hispanic/Latino and 3% were Black.[4]
- There was greater diversity in the CLARITY-AD lecanemab Phase 3 trial, in which 22.5% of US participants were Hispanic/Latino and 4.5% were Black.[3]
- ASCENT-1 and ASCENT-2 are ongoing Phase 1 clinical trials evaluating the safety, tolerability, pharmacokinetics, pharmacodynamics, and immunogenicity of subcutaneously administered PRX012 in participants with AD.
 - PRX012—an investigational, novel, humanized IgG1κ mAb targeting the N-terminus of Aβ—binds with high affinity and avidity to aggregated forms of Aβ, including protofibrils and plaques, and is designed for subcutaneous administration.[10]

Here, we provide an overview of ASCENT-1 and ASCENT-2 clinical trial design and the recruitment efforts Prothena implemented in Phase 1 to help increase ethnic diversity and representation in the studies and better represent the populations at risk for AD.

METHODS

- The ASCENT Phase 1 clinical trial program, consisting of two independent clinical trials, uses a staggered but complementary approach in which a dose level is first explored in a single-ascending dose study (ASCENT-1) followed by a multiple-dose study (ASCENT-2).[11]
 - Select eligibility criteria for study participants are described in **Table 1**.
 - Study objectives and assessment schedules are described in **Figure 1**.

Table 1: Key Eligibility Criteria

Healthy Volunteers: ASCENT-1	Participants With Early AD: ASCENT-1	Participants With Early AD: ASCENT-2
• Aged 20-45 years • No family history of genetically inherited AD or other early onset form of dementia • No contraindication for LP	• Aged 60-85 years • Evidence of AD pathological process, confirmed by amyloid PET visual read	• Aged 55-85 years • Met prespecified CL-based PET imaging criteria
	• Gradual and progressive memory change for ≥6 months • Diagnosis of AD or MCI due to AD according to NIA-AA criteria • MMSE ≥18 • Met MRI criteria for microhemorrhage and/or superficial siderosis at baseline (ARIA-H)	

ARIA-H, amyloid-related imaging abnormality showing microhemorrhages and superficial siderosis; CL, centiloid; LP, lumbar puncture; MCI, mild cognitive impairment; MMSE, Mini-Mental State Examination; MRI, magnetic resonance imaging; NIA-AA, National Institute on Aging and Alzheimer's Association; PET, positron emission tomography.

- ASCENT-1 (PRX012-101) is an ongoing first-in-human, Phase 1, randomized, double-blind, placebo-controlled, single-ascending dose clinical trial of PRX012 administered via subcutaneous injection in healthy volunteers and in participants with early AD. It is being conducted at multiple trial sites in the USA.
- ASCENT-2 (PRX012-102) is an ongoing Phase 1, randomized, double-blind, placebo-controlled multiple-dose clinical trial of PRX012 administered via subcutaneous injection in participants with early AD. Most ASCENT-2 clinical trial sites are in the USA, with additional sites in Europe.
 - Participants are assigned to **Group A** or **Group B** based on apolipoprotein E4 (*APOE4*) genotype to increase representation of homozygous participants, who make up a small proportion of typical AD study populations.
- In both ASCENT-1 and ASCENT-2, demographic information, including reported ethnicity and race, was collected (see **Table 2** for definitions).
- Both clinical trials are ongoing, with demographic analyses including data available as of August 1, 2024.

Table 2: Definitions of Ethnicity and Race in the ASCENT Clinical Trial Program

Ethnicity Categories	Race Categories	
• Hispanic/Latino • Not Hispanic/Latino • Not Reported	• White • Black or African American • Asian • American Indian or Alaska Native	• Native Hawaiian or Other Pacific Islander • Multiple • Other • Not Reported

METHODS (CONTINUED)

- Across the ASCENT clinical trial program, Prothena employed strategic approaches to reach a more ethnically diverse population, including:
 - **Site selection**, which included (1) regions associated with large Hispanic/Latino communities; (2) sites that are motivated to reach a diverse population; and (3) sites that are intentional about hiring diverse personnel (e.g., Spanish-speaking neurologists and staff)
 - Prothena also provided funding to support sites' local advertising campaigns, which enabled them to host events and advertise the clinical trials at local community centers, churches, and memory clinics.
 - **Translation** of **recruitment information** (e.g., website, poster, and flyers) and **participant-facing study materials** (e.g., Informed Consent Forms and Cognitive Scales)
 - Translated materials were provided proactively based on expected patient demographics rather than waiting until a potential participant need was identified.
 - **Recruitment vendors** were asked to propose and execute strategies to reach broader demographics and employ multilingual intake staff and materials.

Figure 1: Trial Design (ASCENT-1 and ASCENT-2)

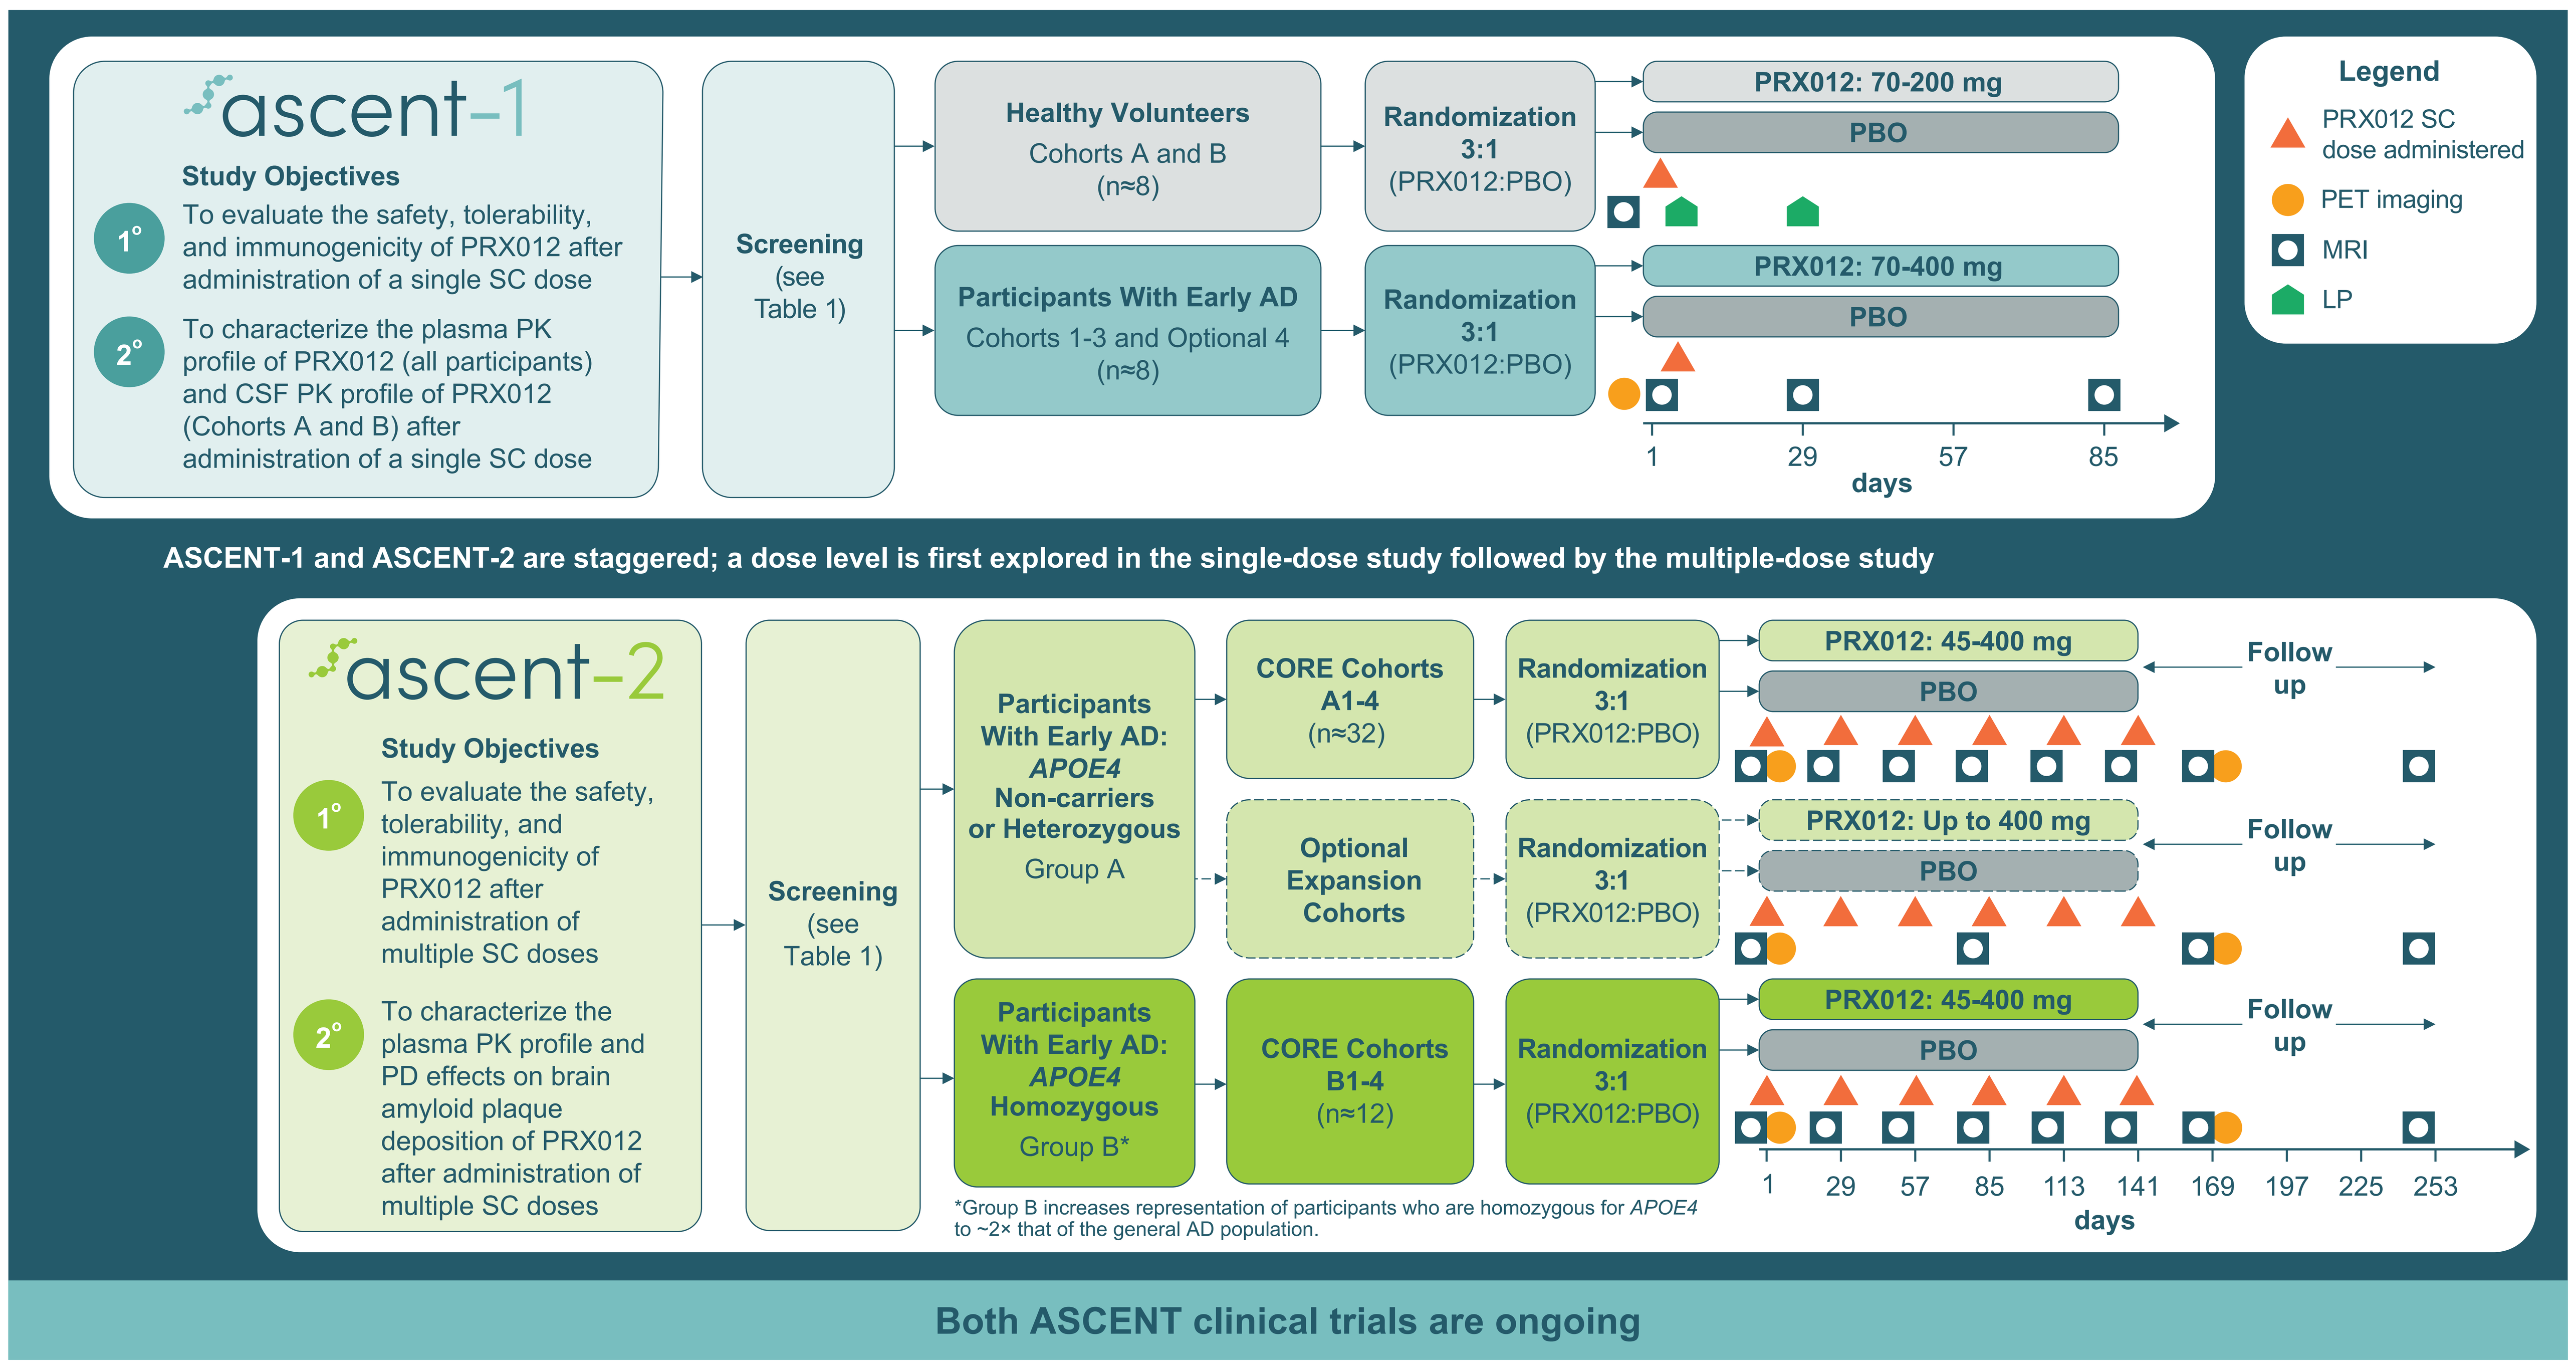
ascent-1
Study Objectives
1°
To evaluate the safety, tolerability, and immunogenicity of PRX012 after administration of a single SC dose
2°
To characterize the plasma PK profile of PRX012 (all participants) and CSF PK profile of PRX012 (Cohorts A and B) after administration of a single SC dose
Screening (see Table 1)
Healthy Volunteers
Cohorts A and B (n≈8)
Participants With Early AD
Cohorts 1-3 and Optional 4 (n≈8)
Randomization 3:1 (PRX012:PBO)
PRX012: 70-200 mg
PBO
PRX012: 70-400 mg
1
29
57
85
days
Legend
PRX012 SC dose administered
PET imaging
MRI
LP
ASCENT-1 and ASCENT-2 are staggered; a dose level is first explored in the single-dose study followed by the multiple-dose study
ascent-2
To evaluate the safety, tolerability, and immunogenicity of PRX012 after administration of multiple SC doses
To characterize the plasma PK profile and PD effects on brain amyloid plaque deposition of PRX012 after administration of multiple SC doses
Participants With Early AD: APOE4 Non-carriers or Heterozygous
Group A
CORE Cohorts A1-4 (n≈32)
Optional Expansion Cohorts
Participants With Early AD: APOE4 Homozygous
Group B*
CORE Cohorts B1-4 (n≈12)
PRX012: 45-400 mg
PRX012: Up to 400 mg
Follow up
1
29
57
85
113
141
169
197
225
253
*Group B increases representation of participants who are homozygous for APOE4 to ~2× that of the general AD population.
Both ASCENT clinical trials are ongoing

CSF, cerebrospinal fluid; PBO, placebo; PD, pharmacodynamic; PK, pharmacokinetic; SC, subcutaneous.

RESULTS: Baseline Demographics

Table 3: Blinded Demographic Data for Participants With Early AD Randomized in ASCENT-1 and ASCENT-2

- Demographic data are presented for the participants with early AD who were randomized in ASCENT-1 and ASCENT-2 (**Table 3**).
- The **ASCENT** clinical program achieved strong enrollment in location(s) with a high percentage of Hispanic/Latino individuals.
- Across the **ASCENT** program, 64% of participants with AD are female, 26% are Hispanic/Latino, and 4% are racially diverse.

	ASCENT-1	ASCENT-2	ASCENT-1 and -2
Age (years)*			
Mean (SD)	74.7 (6.02)	72.6 (6.69)	72.7 (6.65)
Median (Q1, Q3)	77.0 (70, 79)	73.0 (69, 77)	73.0 (69, 78)
Min, Max	61, 85	55, 85	55, 85
Sex			
Male	46.2%	35.5%	36.4%
Female	53.8%	64.5%	63.6%
Ethnicity†			
Hispanic/Latino	15.4%	26.8%	25.8%
Not Hispanic/Latino	84.6%	73.2%	74.2%
Race‡			
White	88.5%	96.2%	95.5%
Black or African American	7.7%	2.6%	3.1%
Asian	3.8%	0.8%	1.0%
Other	0	0.4%	0.3%

Data as of August 1, 2024; n-values for the ASCENT clinical trials have not yet been disclosed.

*Age on date of informed consent; †There were no participants in the "Not Reported" ethnicity category; ‡There were no participants in the following race categories: American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander, Multiple, or Not Reported.

Max, maximum; Min, minimum; Q, quartile; SD, standard deviation.

CONCLUSIONS

- Together, ASCENT-1 and ASCENT-2 are intended to provide data to support and inform design of future late-stage clinical trials that have the potential to position PRX012 as a best-in-class, next-generation, once-monthly subcutaneously delivered mAb for the treatment of AD.
- Similar or greater representation of Hispanic/Latino older adults with AD was observed in the Phase 1 ASCENT clinical trials compared with other recent Phase 3 clinical trials in AD[3,4] and Medicare enrollment.[9]
 - 25.8% of the ASCENT participants are Hispanic/Latino, which is similar to the 22.5% reported in the US population of CLARITY-AD (lecanemab)[3] and higher than the 6% reported in the US population of TRAILBLAZER-ALZ 2 (donanemab)[4] and ~10% of Medicare enrollees.[9]
- The high Hispanic/Latino representation in ASCENT can likely be attributed to proactive targeted recruitment efforts, including intentional site selection, providing up-front financial support for local recruitment campaigns to reach the broader community, and proactively translating materials to reach the Hispanic/Latino population at study start.
- The proportion of Black individuals in ASCENT and other AD clinical trials suggest that targeted efforts to increase Black representation are warranted.
 - 3.1% of the ASCENT participants are Black, which is similar to that reported in the US populations of CLARITY-AD (4.5%)[3] and TRAILBLAZER-ALZ 2 (3%)[4] and less than the ~11% of Medicare enrollees.[9]
- Continued efforts by clinical trial sponsors and sites, such as those described here, may result in improved ethnic and racial diversity in clinical trials for AD, thereby potentially expanding the applicability of study results to a broader and more representative population.

REFERENCES

1. Kumar AP, et al. *Biochem Biophys Res Commun.* 2022;587:58-62; 2. Molineuvo JL, et al. *Acta Neuropathol.* 2018;136:821-853; 3. van Dyck CH, et al. *N Engl J Med.* 2023;388:9-21; 4. Sims JR, et al. *JAMA.* 2023;330:512-527; 5. Schwartz AL, et al. *N Engl J Med.* 2023;388:1252-1254; 6. Canevelli M, et al. *Neurosci Biobehav Rev.* 2019;101:122-128; 7. Franzen S, et al. *Alzheimers Dement.* 2022;18:810-823; 8. Alzheimer's Association. *Alzheimers Dement.* 2024;20:3708-3821; 9. Centers for Medicare & Medicaid Services. CMS Program Statistics – Medicare Total Enrollment. Updated March 6, 2023. Accessed August 15, 2024. https://data.cms.gov/summary-statistics-on-beneficiary-enrollment/medicare-and-medicaid-reports/cms-program-statistics-medicare-total-enrollment; 10. Campbell B, et al. Oral presentation at AD/PD™ 2023; 11. Swanson C, et al. Poster presentation at AAIC 2024.

DISCLOSURES

All authors are current or former employees of Prothena Biosciences Inc and shareholders of Prothena Corporation plc. **HG** was an employee of Prothena Biosciences Inc and shareholder of Prothena Corporation plc at the time of this study. **CS** and **GGK** are inventors on Prothena patents and patent applications. **GGK** has received personal compensation for serving as an officer or member of the Board of Directors for Prothena and for Libra Therapeutics.

ACKNOWLEDGEMENTS

The authors thank the participants and care partners who participated in ASCENT clinical trials, the study sites and investigators, and the Data and Safety Monitoring Board. This study is sponsored by Othair Prothena Limited, Dublin, Ireland, a member of the Prothena Corporation plc group. Editorial support was provided by Joanna Rowell, PhD, AMICULUM, and funded by Prothena Biosciences Inc.

Presented at the Clinical Trials on Alzheimer's Disease (CTAD); October 29-November 1, 2024; Madrid, Spain and online.

*Presenter: Dr. Chad Swanson (chad.swanson@prothena.com)